Survey of Challenges in Access to Diagnostics and Treatment for Neuroendocrine Tumor (NET) Patients (SCAN) – Healthcare Quality Evaluation

Catherine Bouvier[1], Dirk Van Genechten[2], Teodora Kolarova[3], Mark McDonnell[4], Dermot O'Toole[5], Harjit Singh[6], Jie Chen[7], James R. Howe[8], Simron Singh[9], Christine Rodien-Louw[10], Simone Leyden[11], Sugandha Dureja[12]

[1]Neuroendocrine Cancer UK, Leamington Spa, UK, [2]vzw NET & MEN Kanker Belgium, Kortrijk, Belgium, [3]INCA, Boston, US, [4]NET Patient Network, Dublin, Ireland, [5]National Centre for Neuroendocrine Tumours, St. Vincent's University and Department of Clinical Medicine, St. James Hospital and Trinity College, [6]Prince Court Medical Centre 39, Jalan Kia Peng, Kuala Lumpur, Malaysia, [7]The First Affiliated Hospital, Sun Yat-sen University, Guangdong, China, [8]University of Iowa Carver College of Medicine, Iowa City, IA, [9]Sunnybrook Odette Cancer Centre, University of Toronto, Toronto, ON, [10]APTED, Lyon, France, [11]NeuroEndocrine Cancer Australia, Blairgowrie, VIC, AU, [12]CNETS India, New Delhi, India

International Neuroendocrine Cancer Alliance

GOOD SCIENCE
BETTER MEDICINE
BEST PRACTICE

INTRODUCTION

- Neuroendocrine tumors (NETs) are rare and complex neoplasms, affecting various organs, but most commonly the gastrointestinal tract.[1]
- NETs incidence and prevalence is increasing worldwide making it one of the fastest growing classes of cancer.[1]
- The International Neuroendocrine Cancer Alliance (INCA) consists of 26 patient advocacy and research groups and supports NET patients (and their families) by advocating on their behalf to improve diagnosis, care and research.

OBJECTIVE

- SCAN assessed global delivery of NET diagnostics and treatment in terms of:

Awareness | Availability | Quality | Affordability

- This analysis focused on the quality of global delivery of NET healthcare.

METHODS

- During Sept-Nov 2019, NET patients and healthcare professionals (HCPs) completed an online survey.
- The survey was disseminated via social media and NET patient groups' and medical societies' networks.
- The survey was available in 14 languages:
 - Arabic, Bulgarian, English, German, Dutch/Flemish, French, Japanese, Hindi, Italian, Mandarin (Chinese), Portuguese, Russian, Spanish, and Swahili.
- On average, NET patients took 20 minutes and HCPs 11 minutes to complete the questionnaire.

RESULTS

Participant Characteristics

- There were 2795 respondents from 68 countries
 - 436 HCPs of which medical oncologists comprised 24% [103/436]
 - 2359 NET patients/carers: Europe (47% [1102/2359]); North America (31% [727/2359]); Asia (12% [280/2359]); Oceania (9% [200/2359]); South America & Africa (2% [50/2359]).

Diagnosis

- Almost half (44% [1042/2359] of patients were initially misdiagnosed.
- Almost half (46% [1077/2359]) of patients had stage IV NETs or had metastases at diagnosis.
- Mean time to diagnosis was 5 years, lower in Asia and higher in North America (t-test, p<0.0001, Figure 1).

Figure 1: Mean Years to Diagnosis

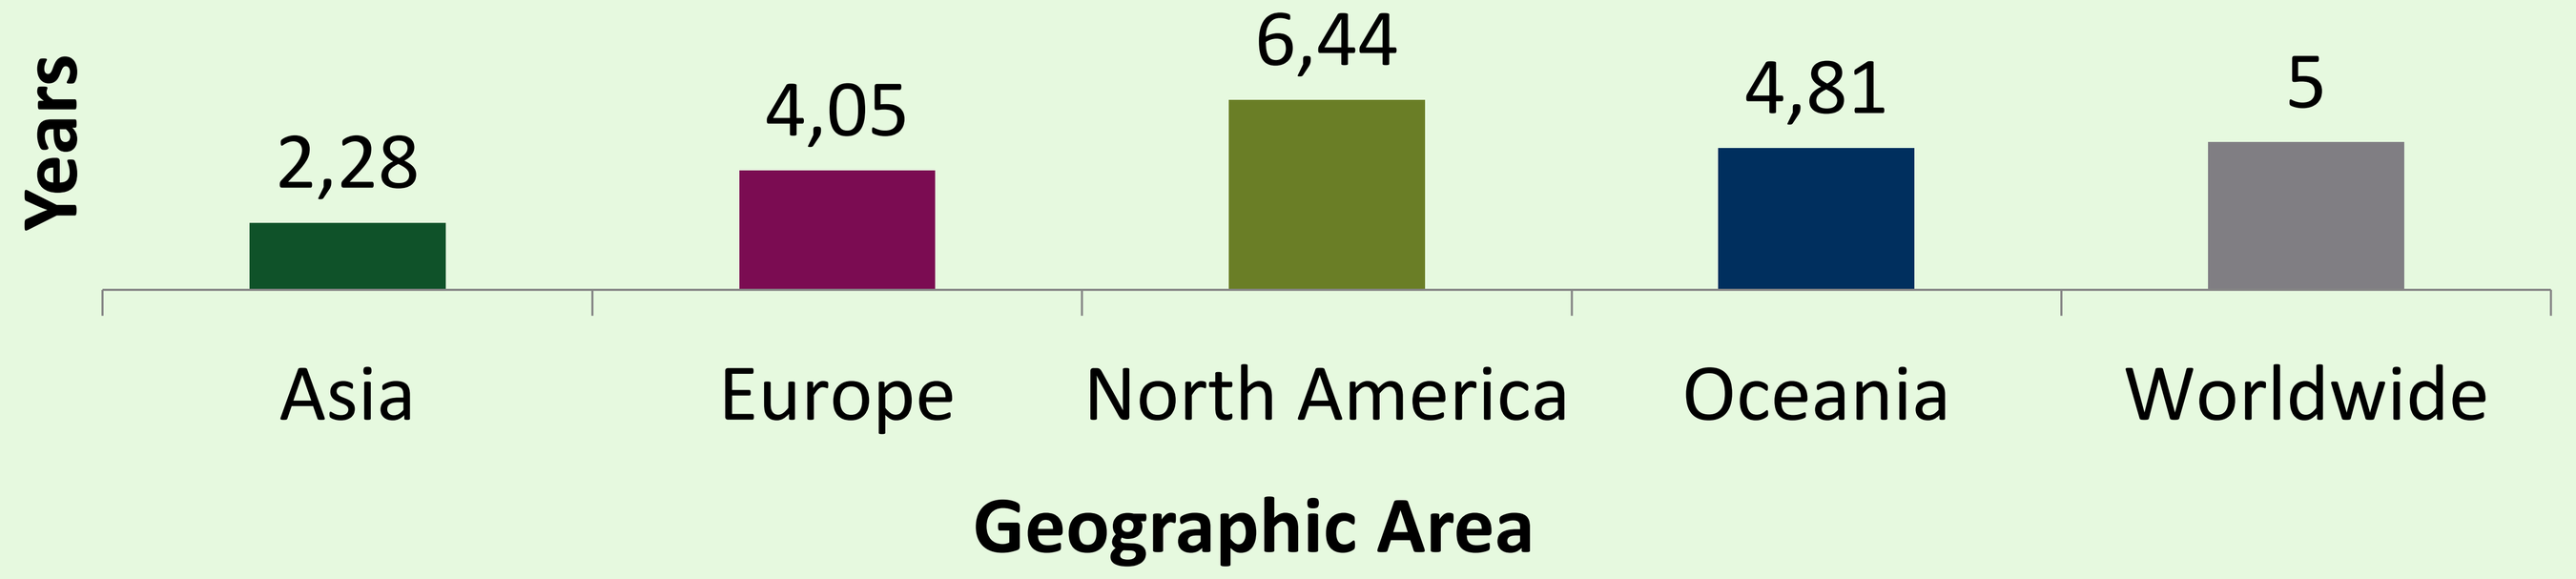
Years
2,28
4,05
6,44
4,81
5
Asia
Europe
North America
Oceania
Worldwide
Geographic Area

Quality of Care

- The quality of healthcare provided was evaluated by NET patients at 3.5 and HCPs/medical oncologists at 3.3/3.1 (5-point Likert scale).

Figure 2: Mean Healthcare Quality Score by health care professionals (HCPs), medical oncologists (MOs) and patients

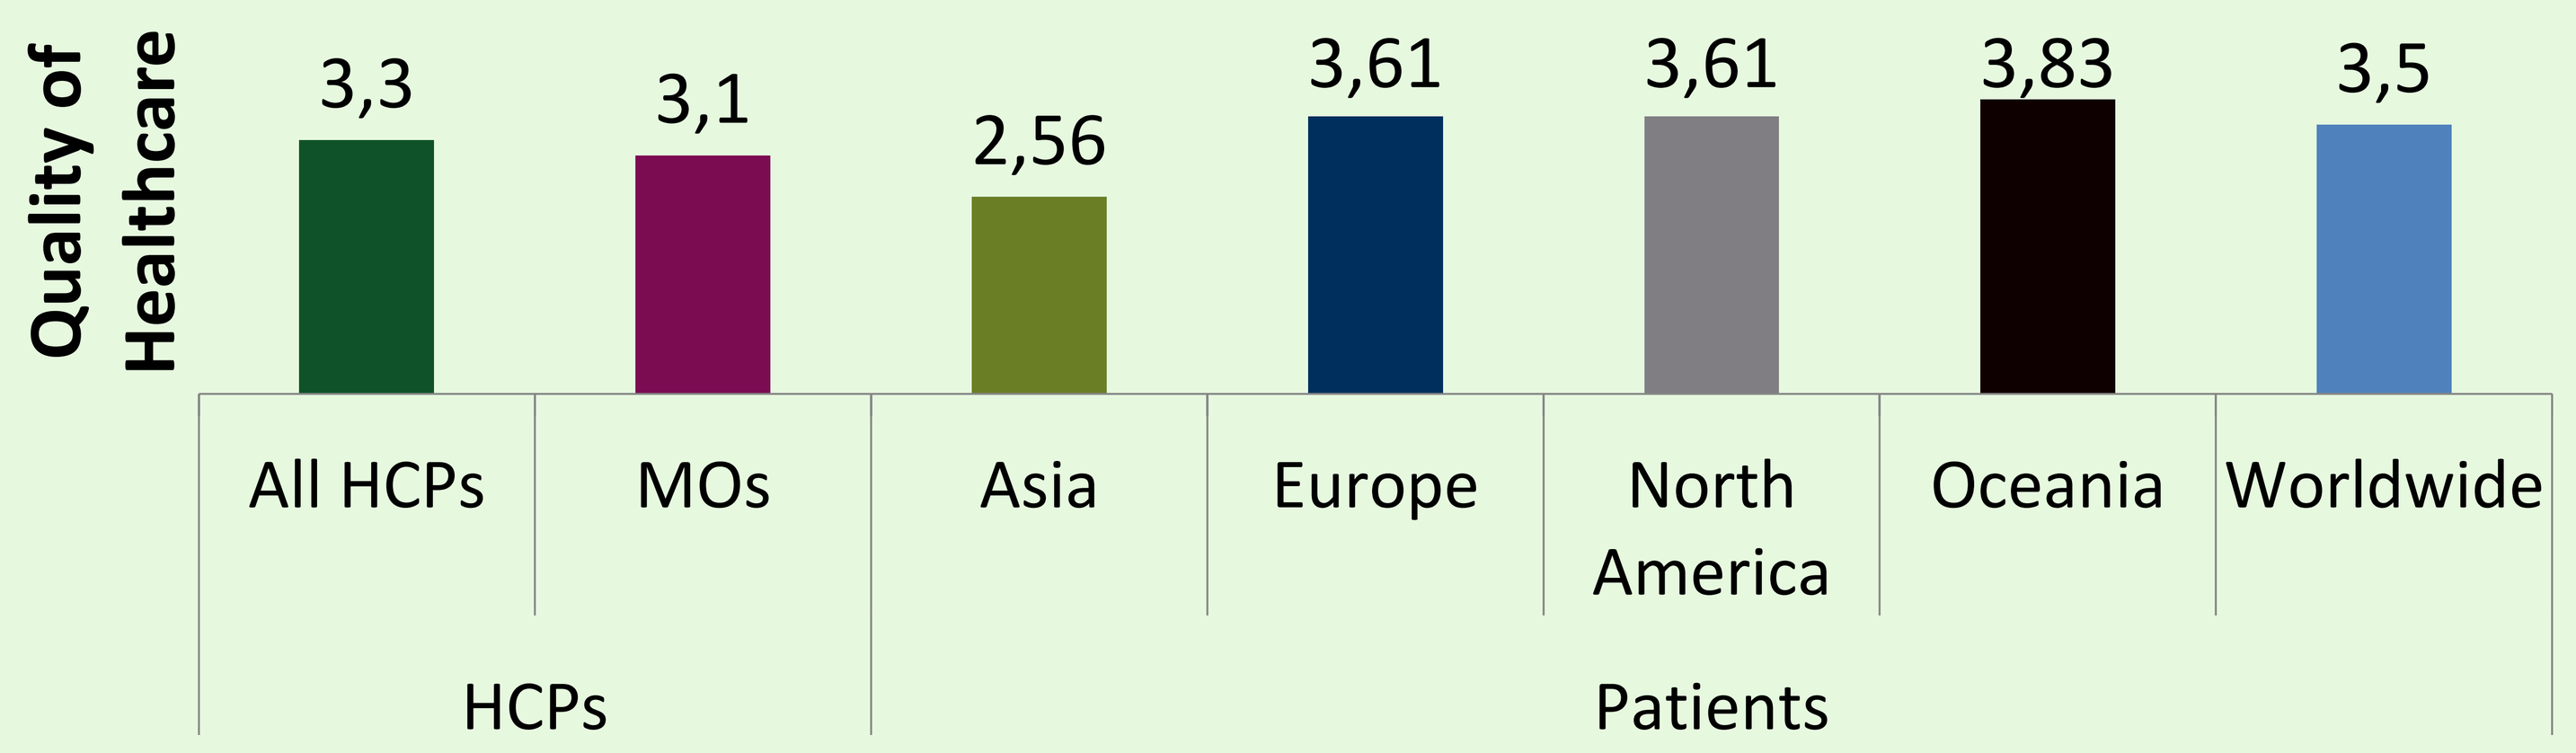
Quality of Healthcare
3,3
3,1
2,56
3,61
3,61
3,83
3,5
All HCPs
MOs
Asia
Europe
North America
Oceania
Worldwide
HCPs
Patients

RESULTS continued

Healthcare Services

- Slightly over half of NET patients were consulted by a NET specialist in the past 12 months (P12M), with the highest rates in Europe and Oceania (All: 53% [1255/2359]; Europe: 61% [668/1102], North America: 48% [350/727], Asia: 43% [120/280], Oceania: 53% [105/200], p<0.0001, Chi-squared, Figure 3).

Figure 3: Patients consulted by a NET specialist in the past 12 months (P12M)

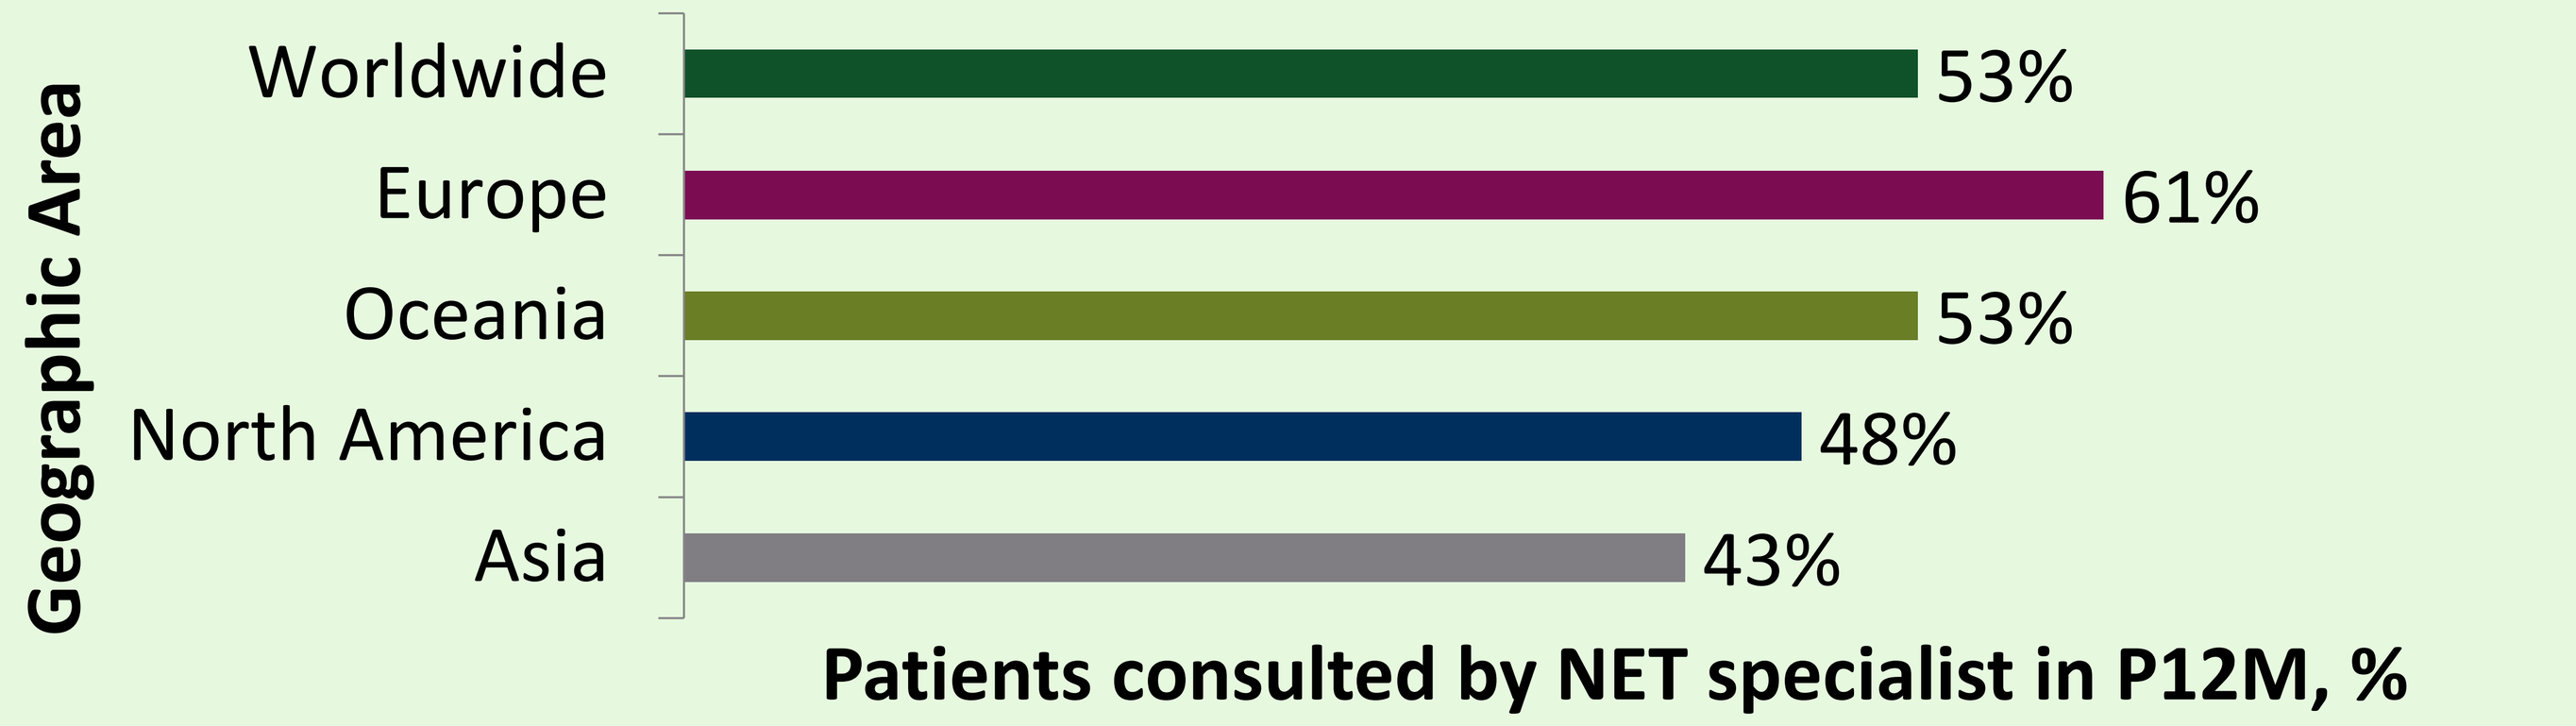
Geographic Area
Worldwide
Europe
Oceania
North America
Asia
53%
61%
53%
48%
43%
Patients consulted by NET specialist in P12M, %

- Only one third of patients were reviewed by a multidisciplinary team in P12M, less in North America and Asia (All: 33% [769/2359]; Europe: 38% [415/1102], North America: 29% [210/727], Asia: 22% [62/280], Oceania: 39% [77/200], p<0.0001).

Figure 4: Patients reviewed by a multidisciplinary team (MDT) in the past 12 months (P12M)

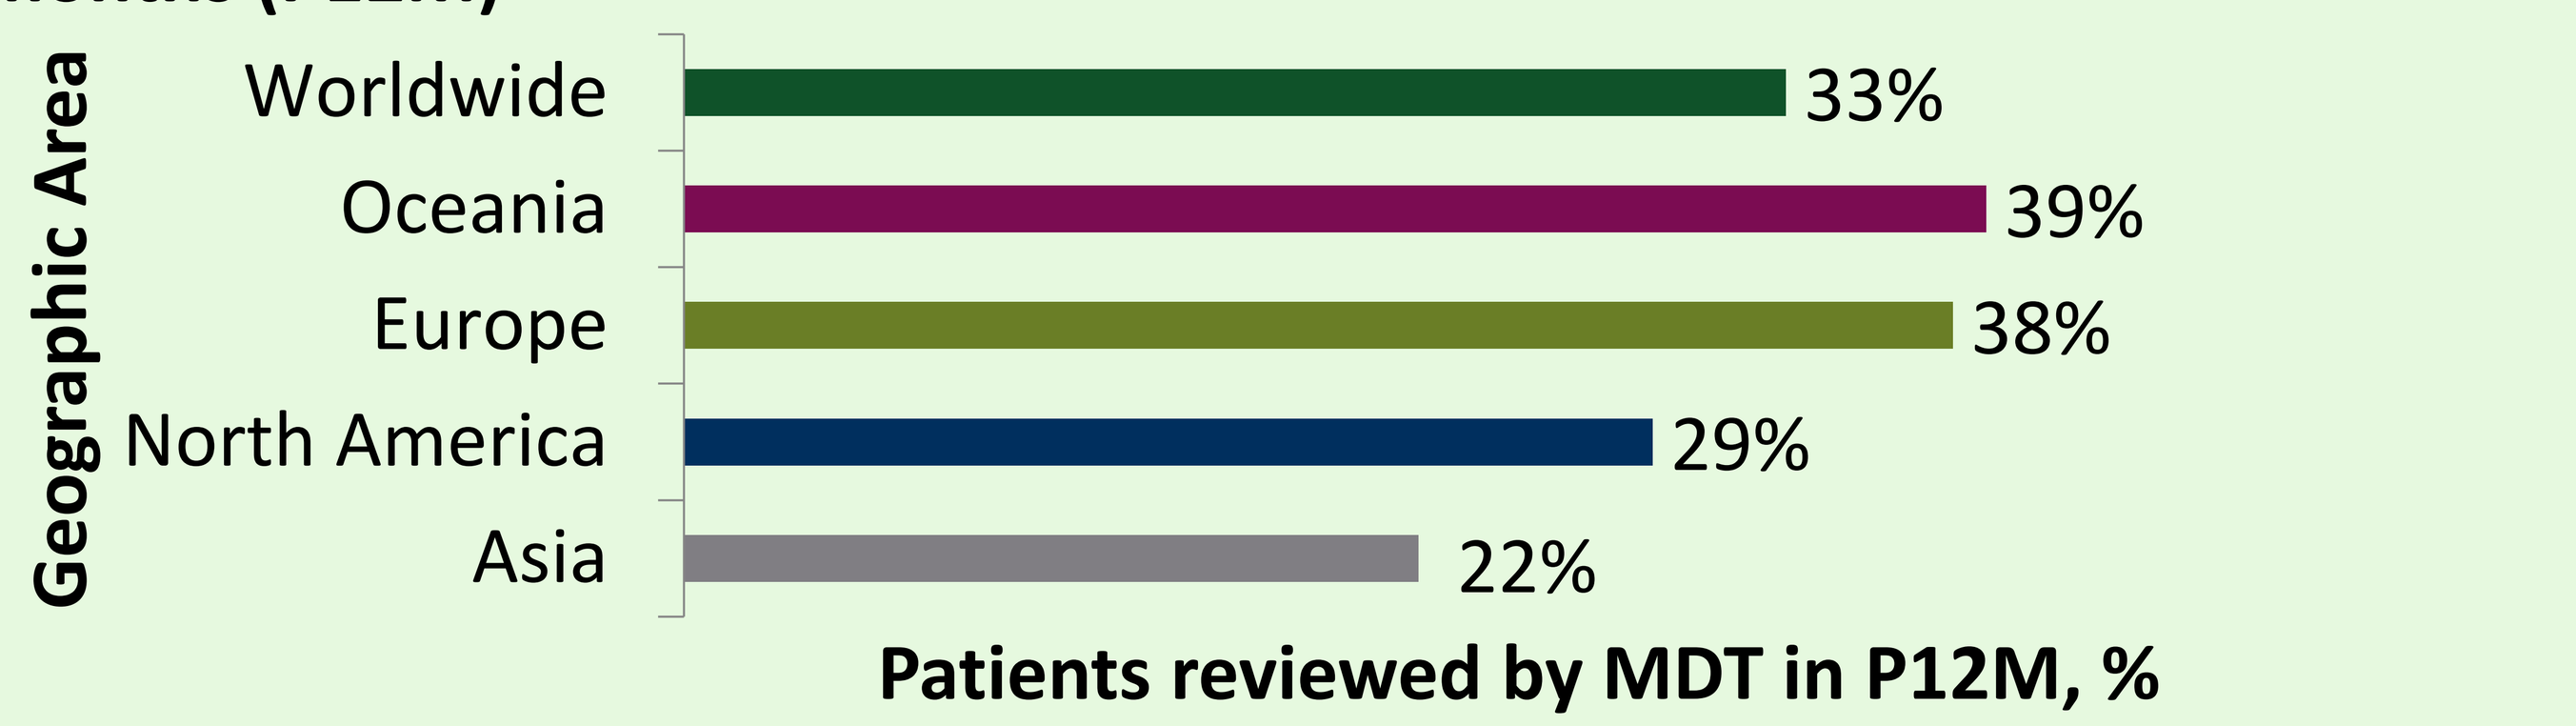
Geographic Area
Worldwide
Oceania
Europe
North America
Asia
33%
39%
38%
29%
22%
Patients reviewed by MDT in P12M, %

- A medical oncologist was the most involved HCP in follow-up in 60% of NET patients, particularly in North America and Oceania (Europe 54% [593/1102] vs. North America 75% [548/727] vs. Asia 34% [95/280], vs. Oceania 75% [150/200] p<0.0001).

Recommendations

- The most common recommendation to improve NET diagnosis and management, given by medical oncologists, was 'better access to NET experts/specialist centers (75% [77/103]).

CONCLUSIONS

- SCAN represents the biggest global compendium of data about NETs extant.
- Delayed NET diagnosis remains a global challenge.
- The delivery of healthcare to NET patients is in need of significant improvement globally to ensure proper and timely diagnosis and treatment.
- SCAN results will help drive forward optimal care and referral pathways to achieve the goal of earlier diagnosis.
- Improving access to NET specialists and multidisciplinary care should be a priority for healthcare systems.
- This survey further contributes to positioning NET research on an equal footing with other cancers of similar prevalence.

REFERENCES

1. Dasari A, et al. JAMA Oncol 2017;3:1335-42.

ACKNOWLEDGEMENTS

INCA would like to thank all its members as well as its partners: ENETS (European Neuroendocrine Tumor Society), NANETS (North American Neuroendocrine Tumor Society), APNETS (Asia-Pacific Neuroendocrine Society), CommNETs (Commonwealth Neuroendocrine Tumor Group), JNETS (Japan Neuroendocrine Tumor Society), CNETS (Chinese Neuroendocrine Tumor Society), UICC (Union for International Cancer Control), EURORDIS (European Organisation for Rare Diseases), NORD (National Organization for Rare Disorders) and ECCO (European Cancer Organisation) and many others for their instrumental support of this global effort.

Strategen Ltd, Winchester, UK provided editorial support to the authors.

FUNDING AND DISCLOSURE

- This study was sponsored by Ipsen, ITM and Novartis.
- The lead author has nothing to declare.

ESMO 2020 Virtual Congress, 18 – 22 September

Correspondence should be sent to **catherine@nc-uk.org**